POLYSACCHARIDES-BASED MATERIALS AS DRUG DELIVERY SYSTEMS

Alexandra Dimofte,[1*] Narcis Anghel,[1] Maria Valentina Dinu,[1] Tudor Boita,[2] Iuliana Spiridon[1]

[1]"Petru Poni" Institute of Macromolecular Chemistry, Iasi, Romania

[2] SC Antibiotice SA, Iasi, Romania

1. Introduction

Alginate and xanthan are well-known polymers that have been used for the development of drug delivery systems.[1]Alginate (Alg) is a polysaccharide comprising mannuronic and guluronic acid residues, obtained either from brown algae or from bacterial sources. It is widely used for drug delivery[2] and in tissue engineering.[3] Xanthan (Xa) is an anionic, high molecular weight polysaccharide produced by the bacterium *Xanthomonas campestris*. It has the ability to form very stiff double-stranded structures.[4] In this work, new drug delivery systems have been developed based on xanthan, xanthan esterified with oleic acid and sodium alginate. Piroxicam (P) has been added into the polysaccharide matrix as anti-inflammatory agent, considering that it is recommended in chronic inflammatory diseases and in controlling postoperative pain.

2. Experimental

2.1. Esterification of xanthan with oleic acid

Esterification of xanthan with oleic acid was performed in the presence of tosyl chloride in methylene chloride. The reaction product (XaAO) was separated by filtration, washed successively with methylene chloride, water and ethanol and then dried at room temperature. FTIR spectra evidenced that the esterification reaction of xanthan took place (Figure 1).

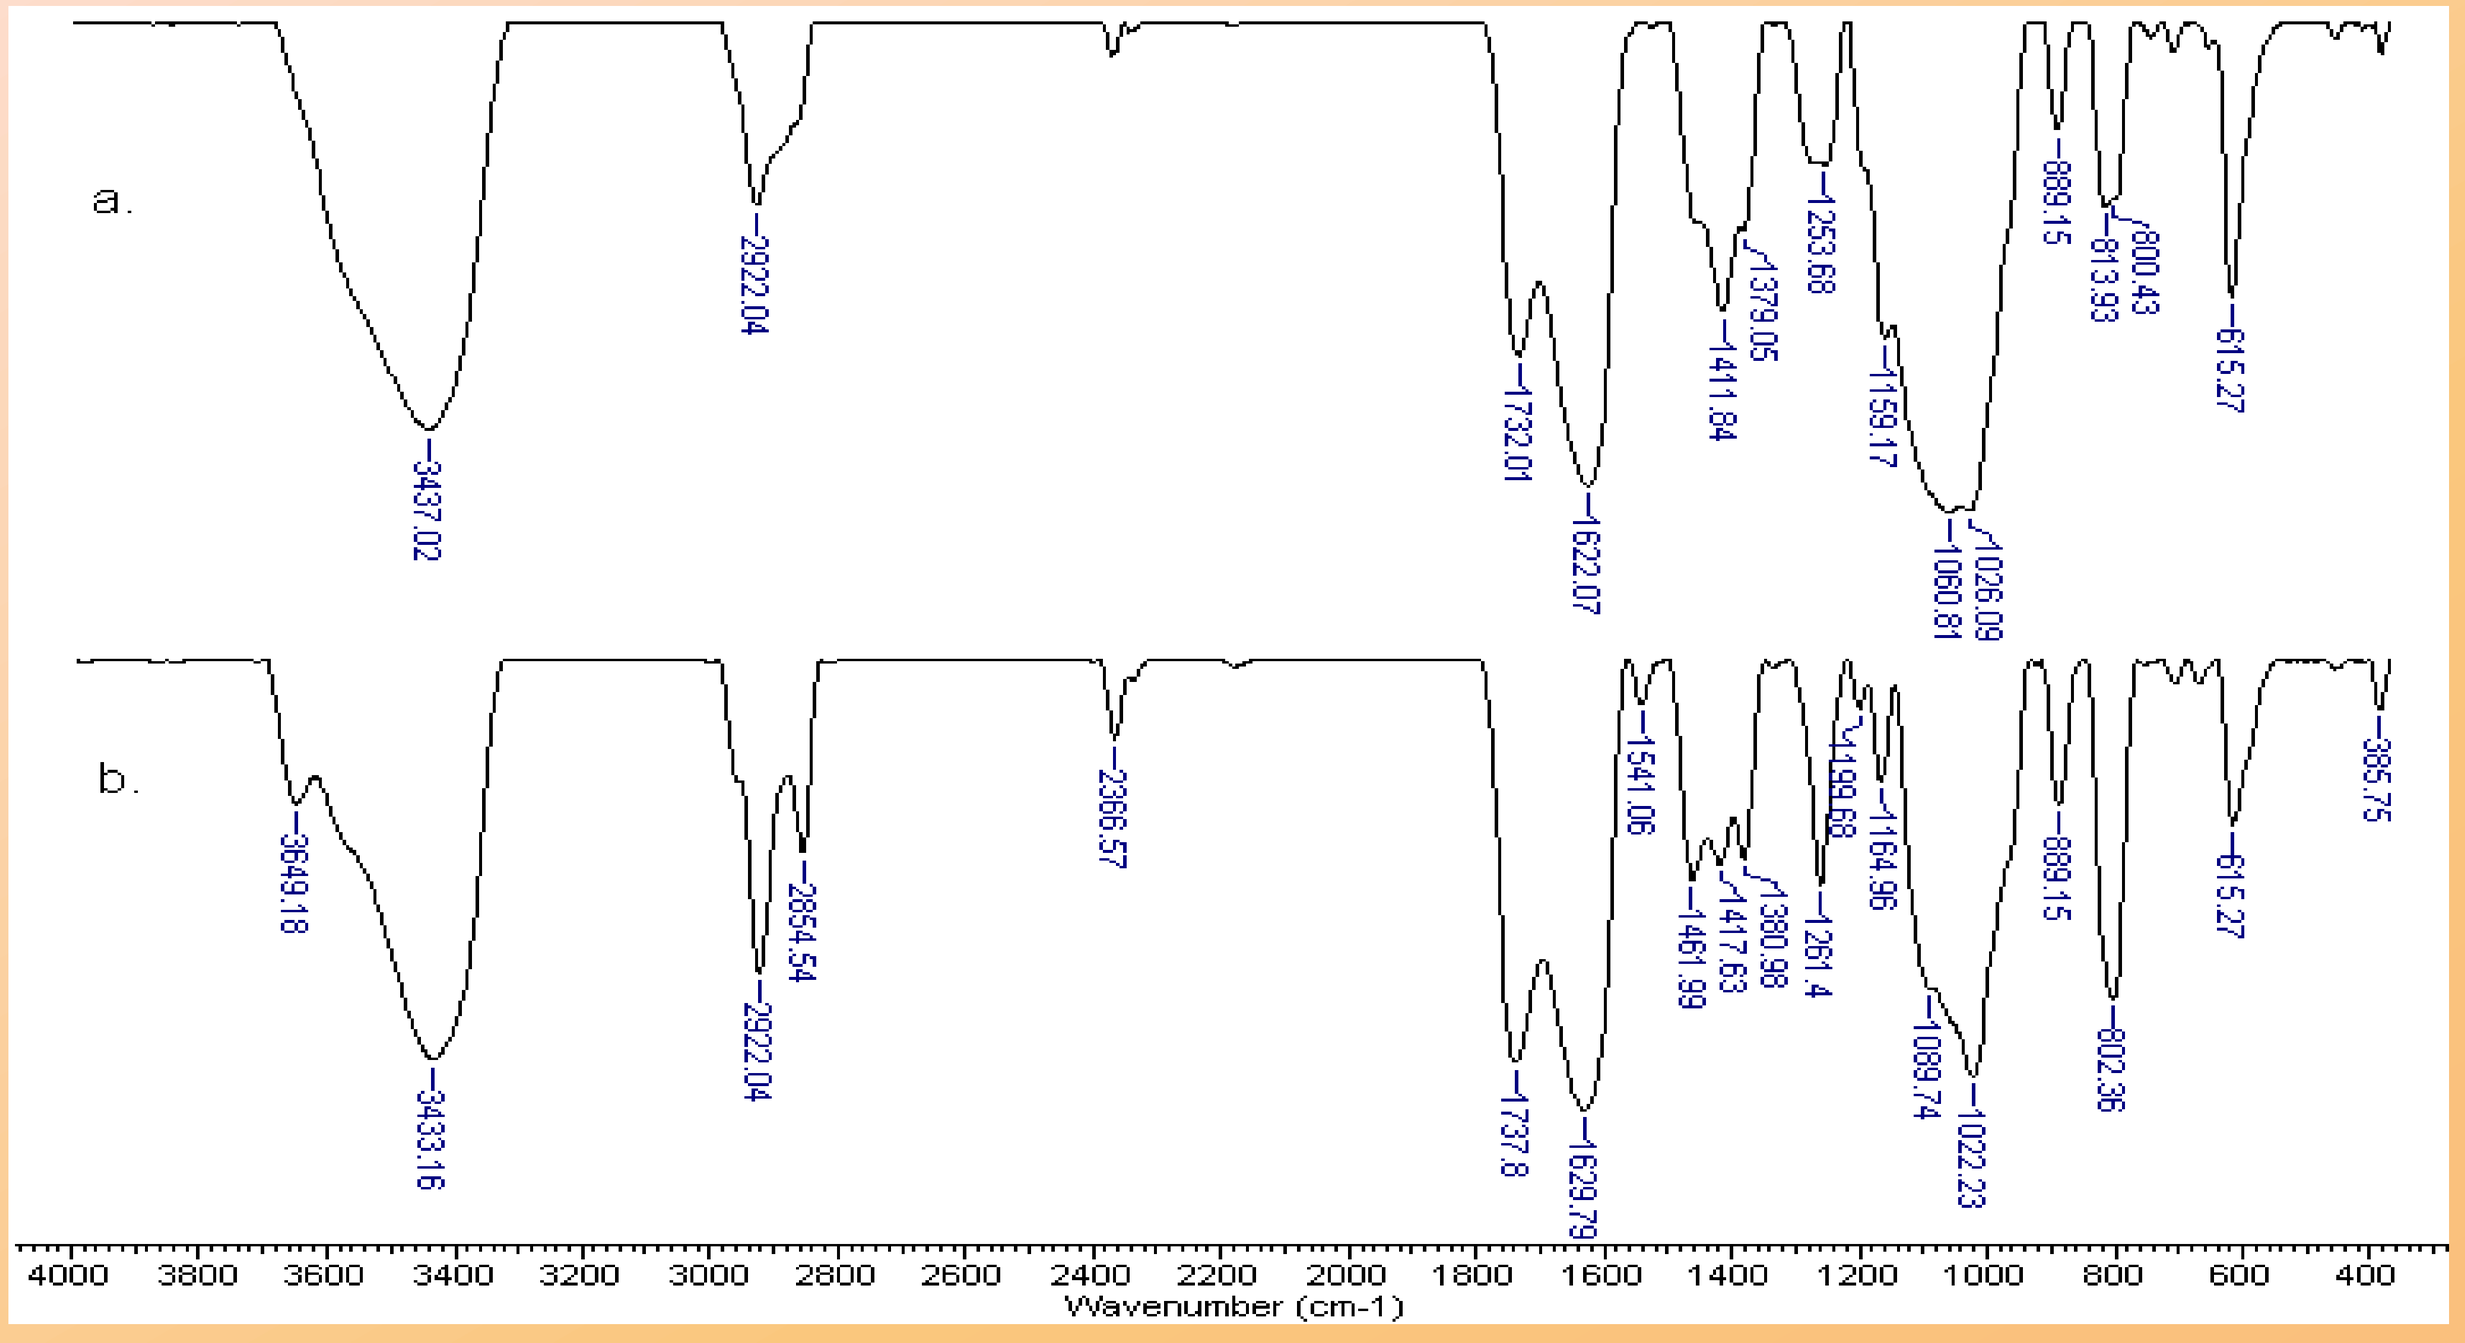
a.
b.
Wavenumber (cm-1)

Figure 1. FTIR spectra for Xa (a) and XaAO (b).

Figure 1b evidenced a decrease in the band intensity characteristic for the hydroxyl groups to 3360 cm^{-1} and an intensification of the stretching vibrations of the –CH2– groups to 2922 cm^{-1} for XaAO. Also, an intensification of the absorption band of the carbonyl group at 1720 cm^{-1} was recorded. These findings confirm the introduction of new ester groups in the xanthan structure.

2.2. Materials and methods

New materials comprising equal amounts of polysaccharides (named Xa-Alg and XaAO-Alg) and 0.05 g of piroxicam (Xa-Alg-P, XaAO-Alg-P) were obtained by freeze-thawing cycles, followed by lyophilization.

FTIR spectra of the materials were recorded using a Vertex 70FTIR spectrometer.

The mechanical properties of materials were evaluated by a Shimadzu Testing Machine EZTest. The SEM images (x200) were taken using a VEGA TESCAN microscope.

UV-Vis measurements were performed on a Jenway 6405 spectrophotometer at 285 nm.

3. Results and discussion

The peaks at 1180 and 1529 cm^{-1} (Figure 2) confirm the presence of piroxicam in the obtained materials.[5]

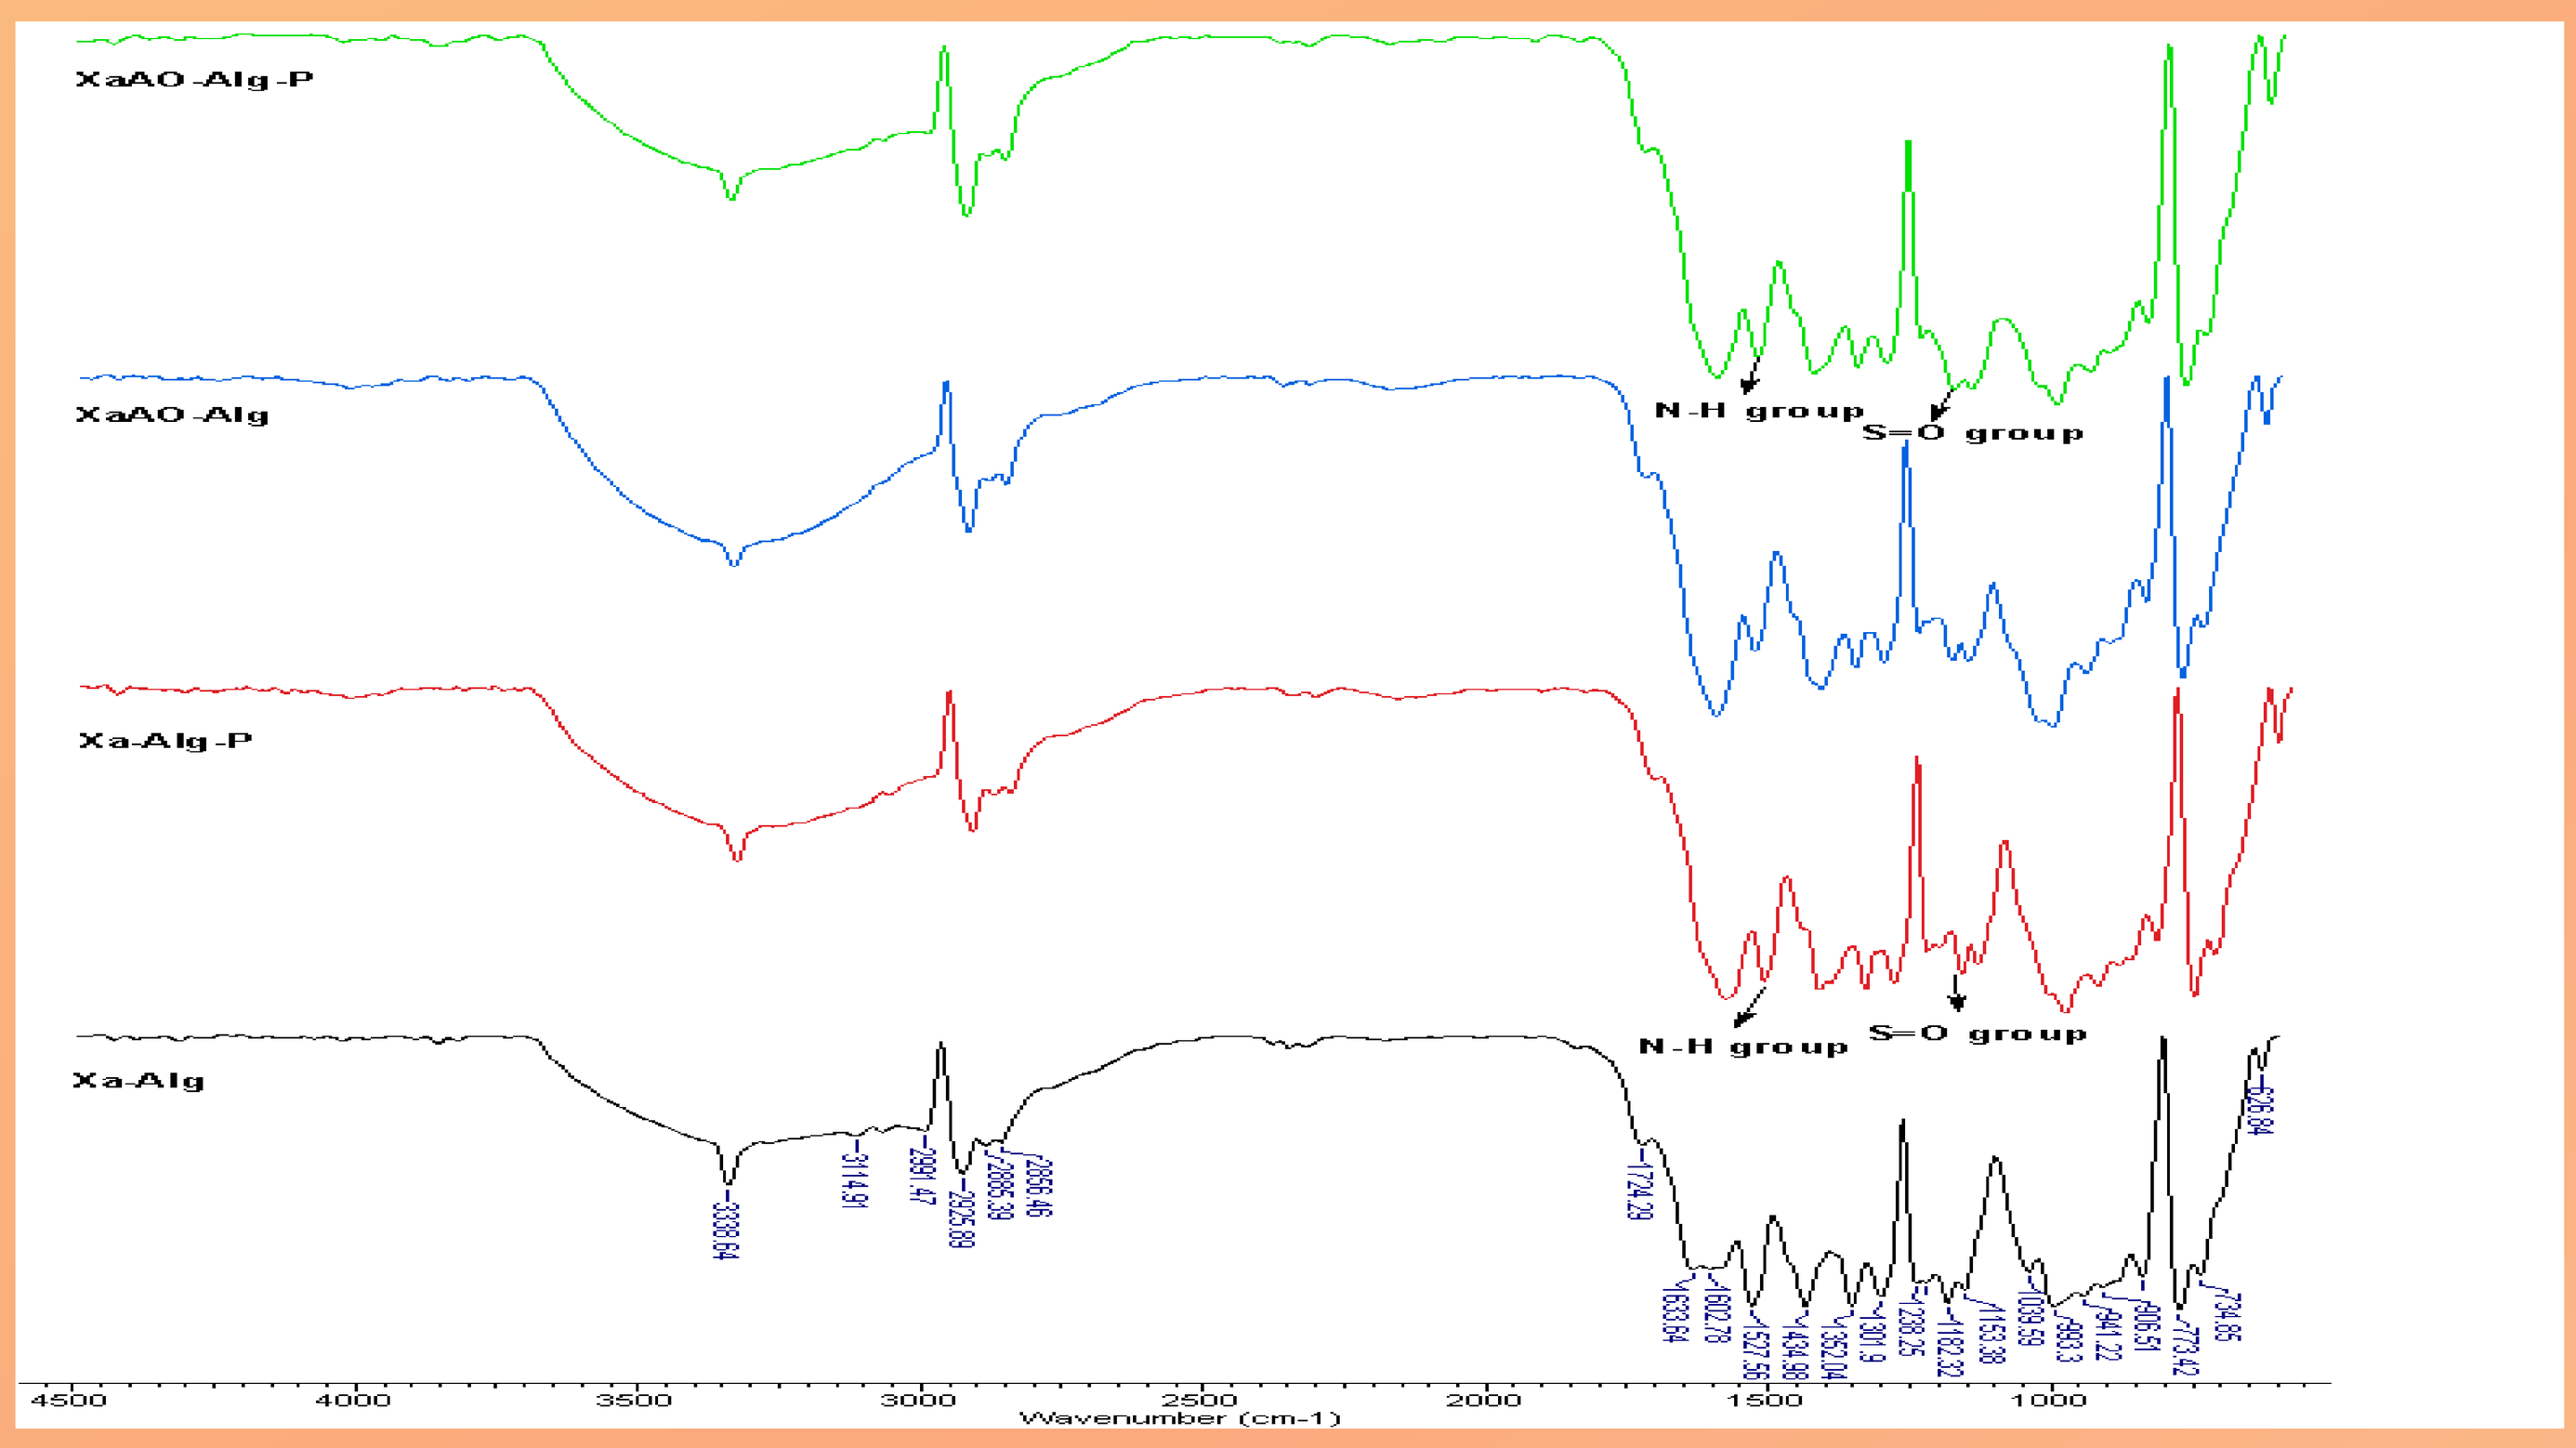
XaAO-Alg-P
XaAO-Alg
Xa-Alg-P
Xa-Alg
N-H group
S=O group
Wavenumber (cm-1)

Figure 2. FTIR spectra of materials based on polysaccharides and piroxicam.

The compressive strength of the material based on modified xanthan was reduced when compared with that of the sample comprising unmodified xanthan (Table 1).

Table 1. Mechanical properties of materials.

Sample	Elastic Modulus*, kPa	R^2	Compressive nominal stress**, kPa	Strain%
Xa-Alg	86.81	0.975	52.47	40.59
Xa-Alg-P	20.21	0.974	27.72	72.51
XaAO-Alg	1.30	0.997	22.40	79.14
XaAO-Alg-P	9.28	0.998	37.83	68.39

Tako *et al*. proposed that intramolecular associations within the xanthan molecule, involving an interaction between an alternate hydroxyl group at C-3 and the adjacent hemiacetal oxygen atom of the D-glucosyl residues with hydrogen bonding, as in cellulose, and between the methyl group of the acetyl residue and the adjacent hemiacetal oxygen atom of the D-glucosyl residue, with van der Waals interaction, are formed.[6] It is possible that the chemical reaction with oleic acid influenced the xanthan capacity to maintain these inter- and intra- molecular associations. When piroxicam was added into the Xa-Alg matrix, the resulted films recorded lower compressive strength (47.16%), with diminished elongation at the break point. It seems that the interactions between the piroxicam and the polysaccharide matrix, at the concentration included into the formed gel network, reduced the intramolecular space inside the backbone, increasing the elasticity of the material. An increment of 68.88% in the compressive strength of the formulation comprising XaAO was recorded.

SEM images (Figure 3) evidence the presence of pores in all materials.

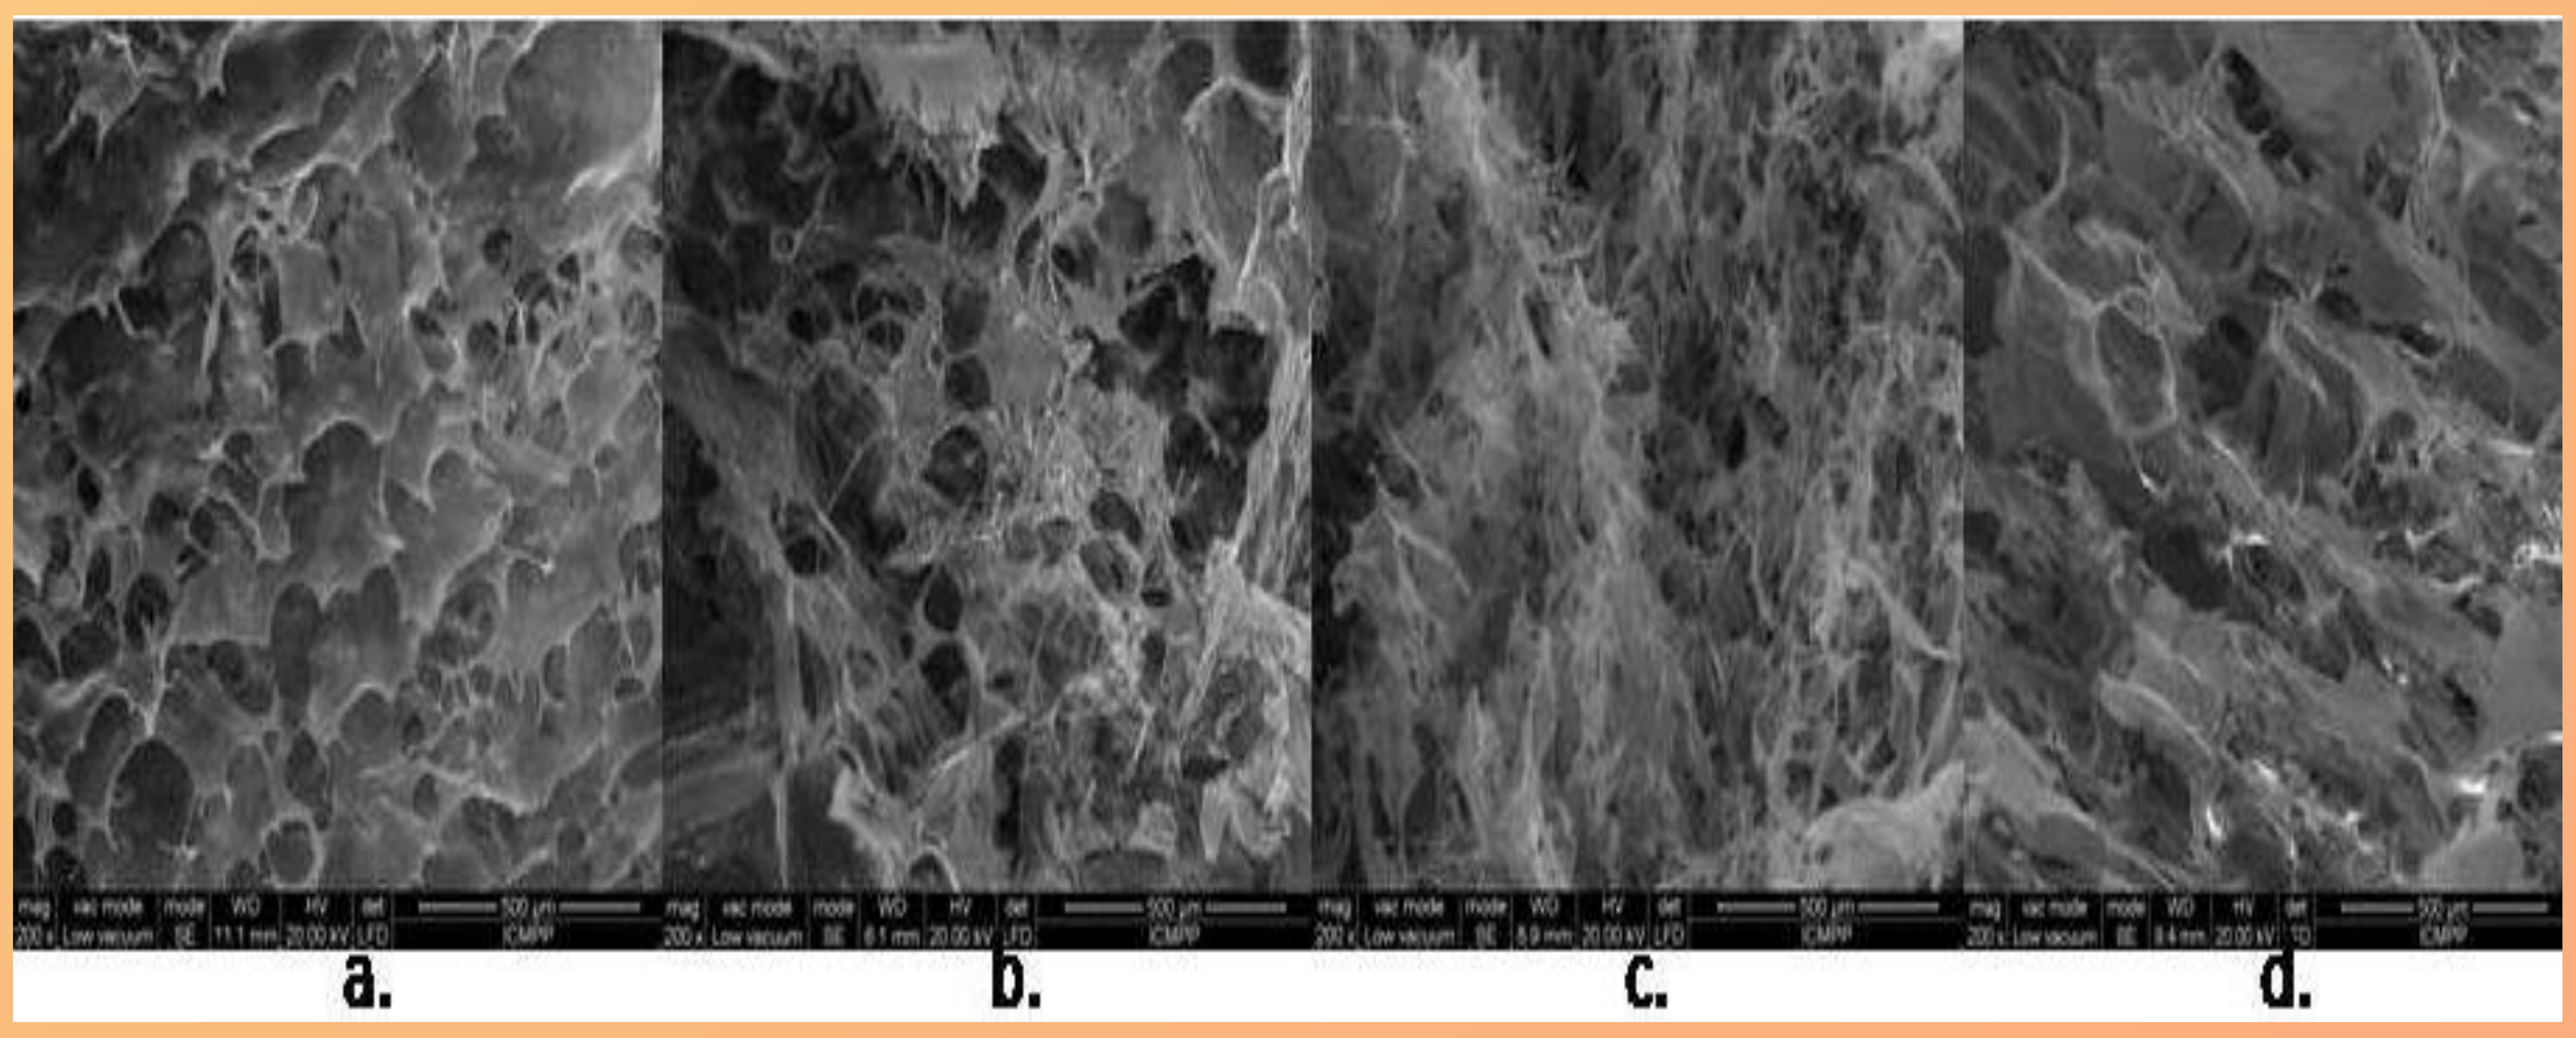
a.
b.
c.
d.

Figure 3. SEM images for Xa-Alg (a), Xa-Alg-P (b), XaAO-Alg (c) and XaAO-Alg-P (d).

The release of active principles from materials is best described by the Korsmeyer-Peppas model.[7]

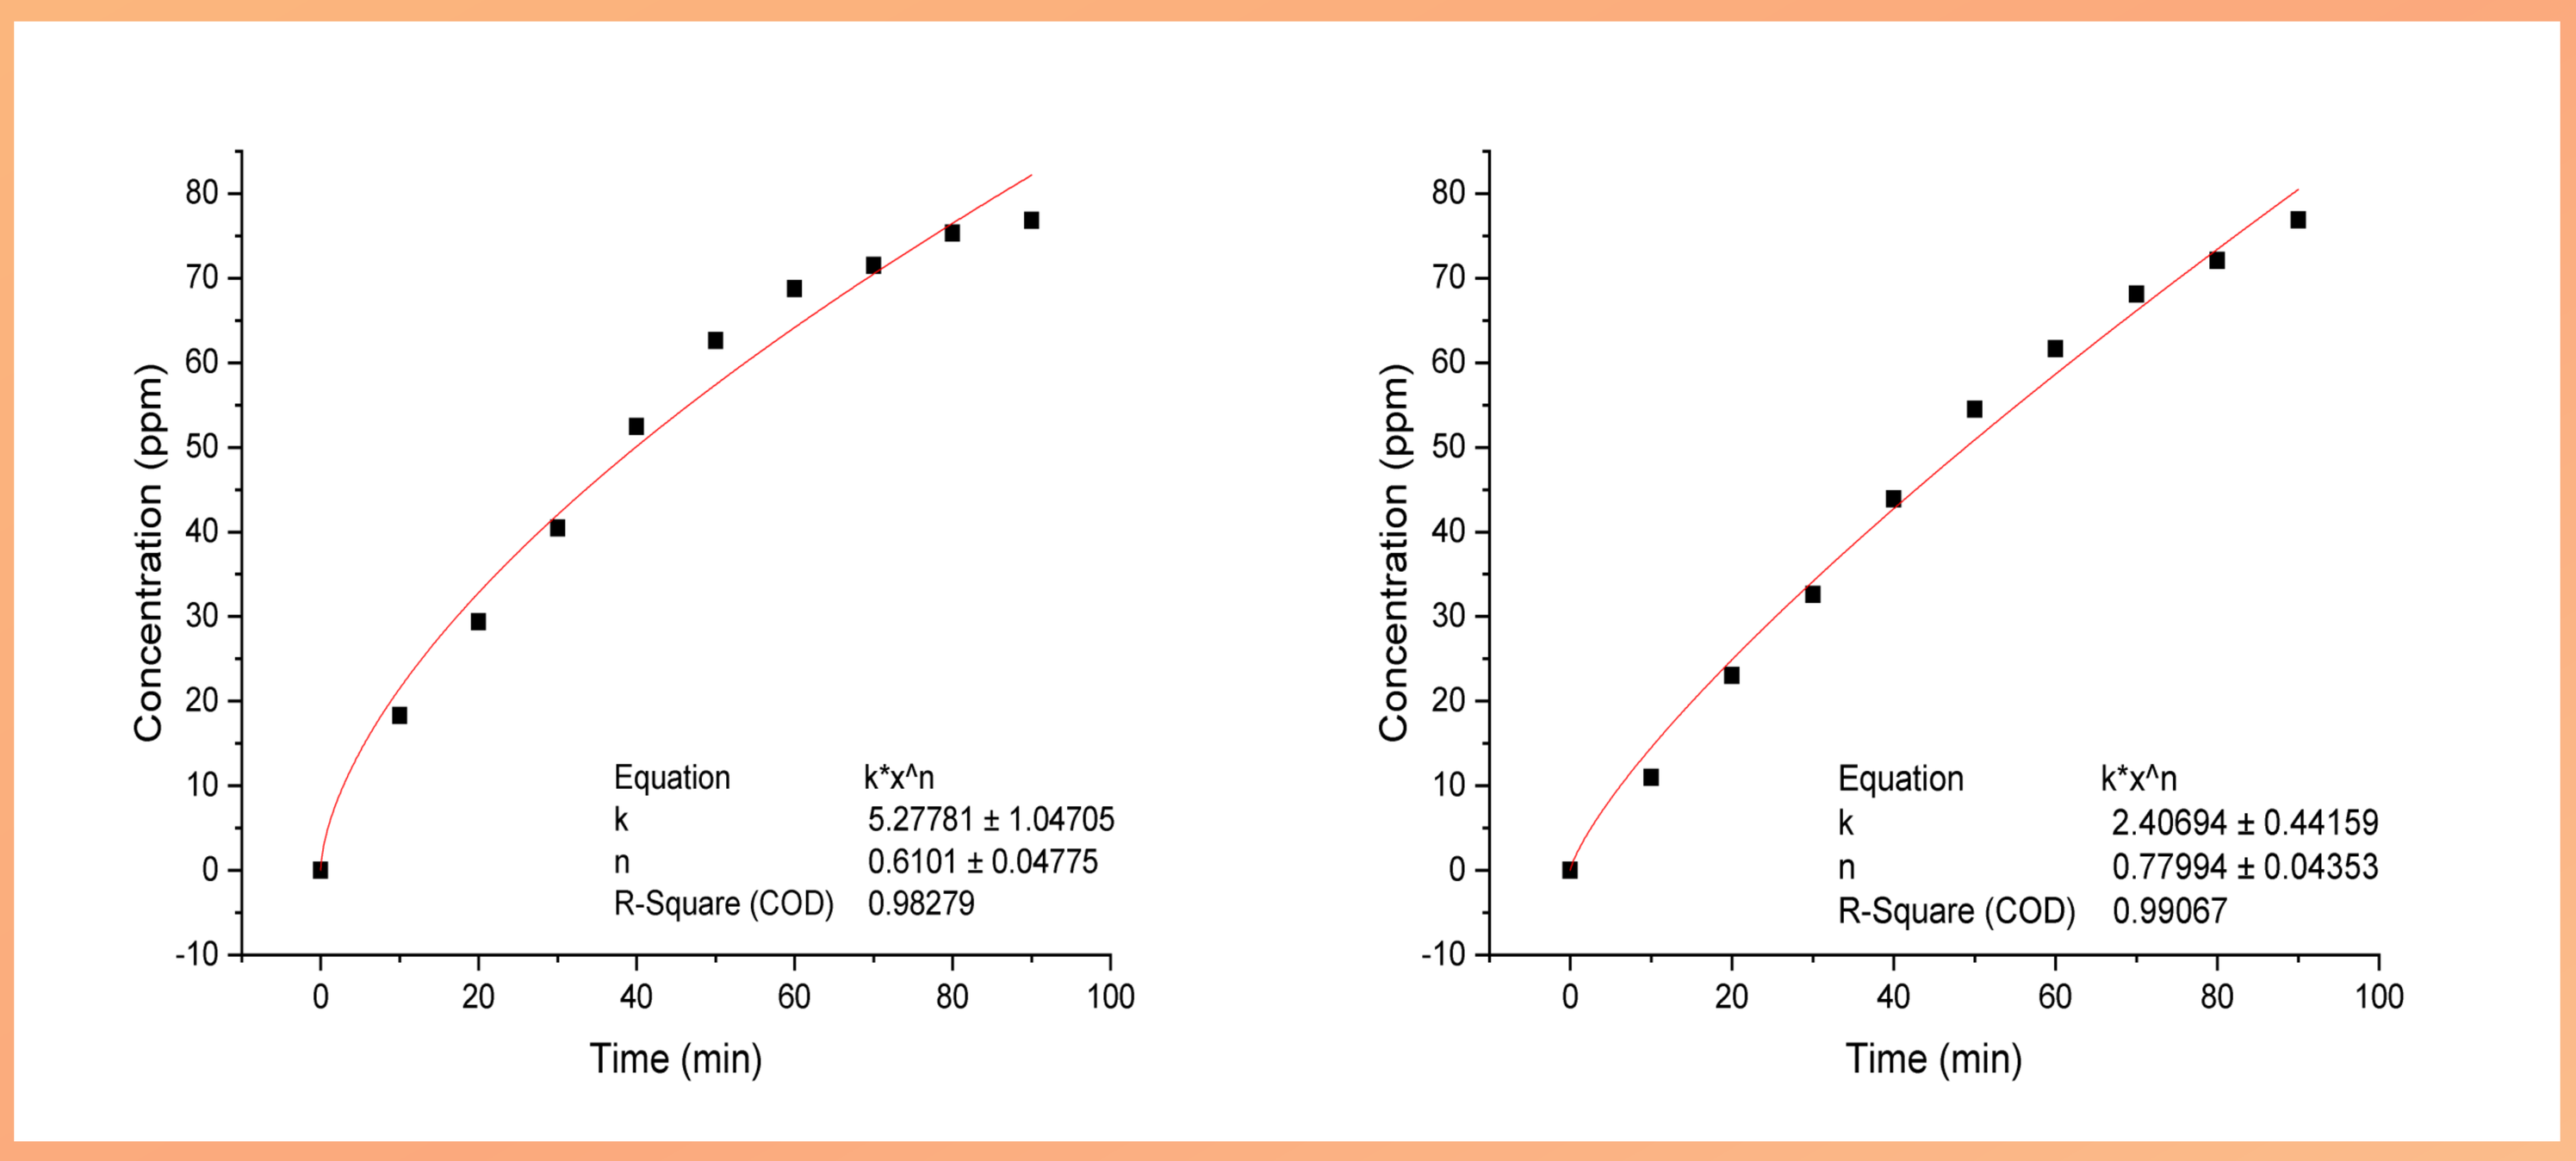
Concentration (ppm)
Time (min)
Equation k*x^n
k 5.27781 ± 1.04705
n 0.6101 ± 0.04775
R-Square (COD) 0.98279
Concentration (ppm)
Time (min)
Equation k*x^n
k 2.40694 ± 0.44159
n 0.77994 ± 0.04353
R-Square (COD) 0.99067

Figure 4. The release of piroxicam from Xa-Alg and XaAO-Alg.

P is released at a slightly faster rate from the material based on Xa (Figure 4), compared with that comprising XaAO (due to the different interaction of the active principle with the polymer matrix).

4. Conclusions

New drug delivery systems based on xanthan and alginate, and containing piroxicam, have been developed and analyzed. The properties of the materials are dependent on the composition of the formulations. When piroxicam was added into the Xa-Alg matrix, a lower compressive strength (47.16%) of the material, along with a decline in elongation at the break, was recorded. An increment of 68.88% in the compressive strength of the formulation comprising XaAO was observed. The strain values decreased after piroxicam addition into the polysaccharide matrix. The release kinetics of piroxicam through the matrix components was explained by the Korsmeyer-Peppas model, with non-Fickian diffusion.

References

[1]F. Freitas, V.D. Alves, M.A.M Reis, *Trends Biotechnol*. 29, 388–398, 2011

[2]H.H. Tønnesen, J. Karlsen, *Drug Dev. Ind. Pharm*. 28, 621–630, 2002

[3]K. Y. Lee, D. J. Mooney, *Prog. Polym. Sci*. 37, 227–258, 2012

[4] O. Aarstad, E. Heggset, I. Pedersen, S. Bjørnøy, K. Syverud, B. Løkensgard Strand, *Polymers* 9, 378, 2017

[5]V. Nikolic, S. Ilic-Stojanovic, L. Nikolic, M. Dobrivoje Cakic, *Hem. Ind*. 68, 107-116, 2014

[6]M. Tako, T. Teruya, T. Tamaki , *Colloid Polym. Sci*. 288, 1161–1166, 2010

[7]M. L. Bruschi, Strategies to modify the drug release from pharmaceutical systems, Ed. Woodhead Publishing, Cambridge, 2015, p. 63-86

Acknowledgements. This work was supported by the project InoMatPol (ID P_36_570, Contract 142/10.10.2016, cod MySMIS: 107464).